AMCP 2022

Financial Burden Associated with Discordance to Intravenous Iron Therapies in US Patients with Iron Deficiency Anemia

Michael Polson, MS, PharmD[1]; Karina Utkina, PharmD[2]; Ryan Bucco, PharmD[2]; Ruslan Horblyuk, PhD, MBA[3]

Magellan Rx MANAGEMENT

[1]Magellan Rx Management • Phoenix, AZ
[2]Pharmacosmos Therapeutics Inc, Morristown, NJ
[3]Aesara, Chapel Hill, NC

Background

Iron deficiency anemia (IDA) affects approximately five million people in the United States and has a significant impact on human health.[1] In a study of 2.3 million health plan members, anemia was observed in 3.5% of the population and they had 54% higher annualized costs per patients than average cost for nonanemic patients.[2] Although oral iron is often viewed as first-line therapy, there is evidence that intravenous (IV) iron is superior, in both efficacy and safety, to oral iron.[3] Several IV iron products are available, including older generation (iron sucrose, iron dextran and sodium ferric gluconate) and newer generation products (ferric derisomaltose, ferric carboxymaltose and ferumoxytol). Newer agents have been shown to better maintain target hemoglobin concentration as well as protect against recurrent events due to anemia.[4,5] Despite the benefits, some payors require failure on older iron products before use of newer iron products in prior authorization processes.

Currently available IV iron replacement regimens requiring multiple infusions may be associated with suboptimal adherence.[6] The need for multiple IV administrations imposes a burden on patients and healthcare delivery system which may result in discordance between actual IV iron administrations and product label. There may be a cost associated with this discordance and the resultant costs outweigh the difference in price between the older and newer products.

Objective

This study seeks to quantify the burden of discordance to IV iron therapy and associated economic consequences.

Methods

- This is a retrospective study using administrative claims data from patients who are enrolled in a commercial insurance program with a regional health plan. Claims incurred between January 2016 and December 2019 were considered for inclusion in the study.
- A course of IV iron therapy is defined as all infusions that occur within six weeks of the initial infusion.
- Discordance to therapy is defined as having received less than 1 gram of iron over a course of therapy.
- Statistical methods employed in this study include chi-square tests for categorical variables, t-tests for comparisons of continuous variables among cohorts, and logistic regression in accounting for differences in the population characteristics.
- Inclusion Criteria
 - o Therapy with an IV iron product
 - » First IVI administration date is the index date
 - o A diagnosis of IDA in the baseline period (12 months prior to initial IV iron infusion)
 - o Age ≥ 18 on index date
 - o Continuous enrollment with both medical and pharmacy coverage in the baseline period as well as the 12 months (inclusive) following the index date (follow-up period)
- Exclusion Criteria
 - o A claim for hemodialysis at any point during the study period

Discordance to intravenous iron therapy in patients with iron deficiency anemia is associated with **higher overall cost of care.**

Table 1a – Demographics by Generation

		All	Older	Newer
n		24,736	10,546	14,190
Age Group	18-30	2,470 (10.0%)	1,143 (10.8%)	1,327 (9.4%)
	31-40	5,152 (20.8%)	2,394 (22.7%)	2,758 (19.4%)
	41-50	8,123 (32.8%)	3,369 (31.9%)	4,754 (33.5%)
	51-64	7,996 (32.3%)	3,234 (30.7%)	4,762 (33.6%)
	65+	995 (4.0%)	406 (3.8%)	589 (4.2%)
Age (mean)		46.33 (11.81) [47; 18 - 97]	45.69 (11.90) [46; 18 - 92]	46.80 (11.73) [47; 18 - 97]
Gender	F	21,099 (85.3%)	8,990 (85.2%)	12,109 (85.3%)
	M	3,637 (14.7%)	1,556 (14.8%)	2,081 (14.7%)
Deyo-Charlson Comorbidity Index	0	12,373 (50.0%)	5,326 (50.5%)	7,047 (49.7%)
	1	4,666 (18.9%)	1,922 (18.2%)	2,744 (19.3%)
	2	2,555 (10.3%)	1,069 (10.1%)	1,486 (10.5%)
	3+	5,142 (20.8%)	2,229 (21.1%)	2,913 (20.5%)
	mean	1.59 (2.53) [0; 0 - 20]	1.59 (2.51) [0; 0 - 20]	1.59 (2.55) [1; 0 - 18]

Table 1b – Demographics by Concordance

		All	Concordant	Discordant
n		24,736	16,699	8,037
Age Group	18-30	2,470 (10.0%)	1,562 (9.4%)	908 (11.3%)
	31-40	5,152 (20.8%)	3,426 (20.5%)	1,726 (21.5%)
	41-50	8,123 (32.8%)	5,566 (33.3%)	2,557 (31.8%)
	51-64	7,996 (32.3%)	5,471 (32.8%)	2,525 (31.4%)
	65+	995 (4.0%)	674 (4.0%)	321 (4.0%)
Age (mean)		46.33 (11.81) [47; 18 - 97]	46.53 (11.64) [47; 18 - 97]	45.90 (12.15) [46; 18 - 92]
Gender	F	21,099 (85.3%)	14,272 (85.5%)	6,827 (84.9%)
	M	3,637 (14.7%)	2,427 (14.5%)	1,210 (15.1%)
Deyo-Charlson Comorbidity Index	0	12,373 (50.0%)	8,386 (50.2%)	3,987 (49.6%)
	1	4,666 (18.9%)	3,219 (19.3%)	1,447 (18.0%)
	2	2,555 (10.3%)	1,746 (10.5%)	809 (10.1%)
	3+	5,142 (20.8%)	3,348 (20.0%)	1,794 (22.3%)
	mean	1.59 (2.53) [0; 0 - 20]	1.56 (2.50) [0; 0 - 20]	1.66 (2.59) [1; 0 - 18]

Count measures: n (%)
Continuous measures: mean (standard deviation) [median; min - max]

Table 2 – Discordance to Therapy by Generation

	All	Older	Newer	p-value
n	24,736	10,546	14,190	
IV Iron Discordance	8,037 (32.5%)	5,781 (54.8%)	2,256 (15.9%)	< 0.0001

Table 3 – Logistic Regression Model for Discordance by Generation

Parameter	Point Estimate	95% Confidence Limits		p-value
Generation (Older)	6.404	6.034	6.798	<.0001
Celiac Disease	1.422	1.116	1.812	0.004
Crohn's Disease	1.398	1.214	1.611	<.0001
Colon Cancer	1.401	1.145	1.715	0.001
Pregnancy	1.220	1.075	1.383	0.002

Variables for inclusion in model were chosen by stepwise selection.

Table 4 – Total Cost of Care by Generation (in US dollars)

	All	Older	Newer	p-value
Total Cost of Care	36,552	38,164	35,353	0.001

Table 5 – Total Cost of Care by Generation and Concordance (in US dollars)

	Older	Newer
Concordant	35,675	33,873
Discordant	40,217	43,185

Results

- There are 24,736 patients included in the study.
- Baseline demographics, including age, gender, and comorbidity index, were similar between the patients who received older versus newer generation products (Table 1a) and patients who were concordant versus discordant (Table 1b).
- Discordance to IV iron therapy overall is 33%. Patients who received newer generation products are less discordant to therapy (16%) than were patients who received older generation products (55%) (Table 2).
- After adjusting for baseline comorbidities patients who received older generation therapies are over six times as likely to be discordant to therapy than as those who received newer generation products (Table 3).
- In general patients who received newer generation therapies had a lower total cost of care than patients who received older generation therapies (Table 4).
- The lowest cost patients included in the study were those who were concordant to newer generation iron products while the highest cost was seen in patients discordant to newer generation products (Table 5).

Conclusion/Discussion

In this study discordance to the older generation products was significantly higher than newer generation products. Patients who were concordant to therapy and on a newer generation product had the lowest total cost of care, suggesting that overall cost of care is not necessarily proportional to the purchase price of the chosen IV iron replacement therapy. This trend continues to hold true after adjusting for patient comorbidity burden. Optimizing concordance to IV iron therapy may lead to lower total cost of care in the IDA population. There remains an unmet need to further reduce the rate of discordance to therapy.

Disclosures

This research was produced by Magellan Rx Management with funding from Pharmacosmos Therapeutics Inc.

References

1. Miller JL. *Cold Spring Harb Perspect Med* 2013;3:a011866
2. Nissenson AR, Wade S, Goodnough T, Knight K, Dubois RW. Economic Burden of Anemia in an Insured Population. *J Manag Care Pharm.* 2005;11(7):565-574. doi:10.18553/jmcp.2005.11.7.565
3. Auerbach M, Deloughery T. Single-dose intravenous iron for iron deficiency: a new paradigm. *Hematol Am Soc Hematol Educ Program.* 2016;2016(1):57-66. doi:10.1182/asheducation-2016.1.57
4. Wise J. High dose iron regimen improves outcomes in dialysis patients, UK study finds. BMJ. 2018;363. doi:10.1136/bmj.k4581
5. Macdougall IC, White C, Anker SD, et al. Intravenous Iron in Patients Undergoing Maintenance Hemodialysis. N Engl J Med. 2019;380(5):447-458. doi:10.1056/NEJMoa1810742
6. IPD Analytics. Hematologic: Iron Deficiency Anemia/Iron Replacement. Accessed March 31, 2021. https://www.ipdanalytics.com/